OmniCAR, a universal CAR T cell therapy utilizing covalent SpyCatcher/SpyTag binding to target multiple antigens in different tumors

Kevin Sek[1,2], Jun-Ming Tong[1,2], Christina Scheffler[1,2], Jasmine Li[1,2], Steven Yatomi-Clarke[3], Rebecca Lim[3], Philip K, Darcy[1,2]

1. Peter MacCallum Cancer Centre, Melbourne, Victoria, Australia; 2. Sir Peter MacCallum Department of Oncology, University of Melbourne, Victorian Comprehensive Cancer Centre; 3. Prescient Therapeutics Limited, Melbourne, Australia

<u>Universal CAR T cell therapy</u>

CAR T cell therapies are associated with unique complications that can arise which may limit their effectiveness. Furthermore, conventional CAR T cell therapy have thus far shown limited efficacy to treat solid tumours, due to factors such as the immunosuppression in the tumour microenvironment (TME), inefficient T cell trafficking and heterogeneity of antigen expression. In the context of both solid and haematological malignancies, relapse is common due to tumour escape.

<u>OmniCAR platform</u>

The OmniCAR universal immune receptor is comprised of two discrete components, that when combined forms an unbreakable covalent bond to act as an immunologic bridge to target tumour antigens, eliciting an antigen specific T cell response.

'Arming' OmniCAR T cells against multiple tumor antigens through rapid and spontaneous covalent SpyTag/SpyCatcher bond formation

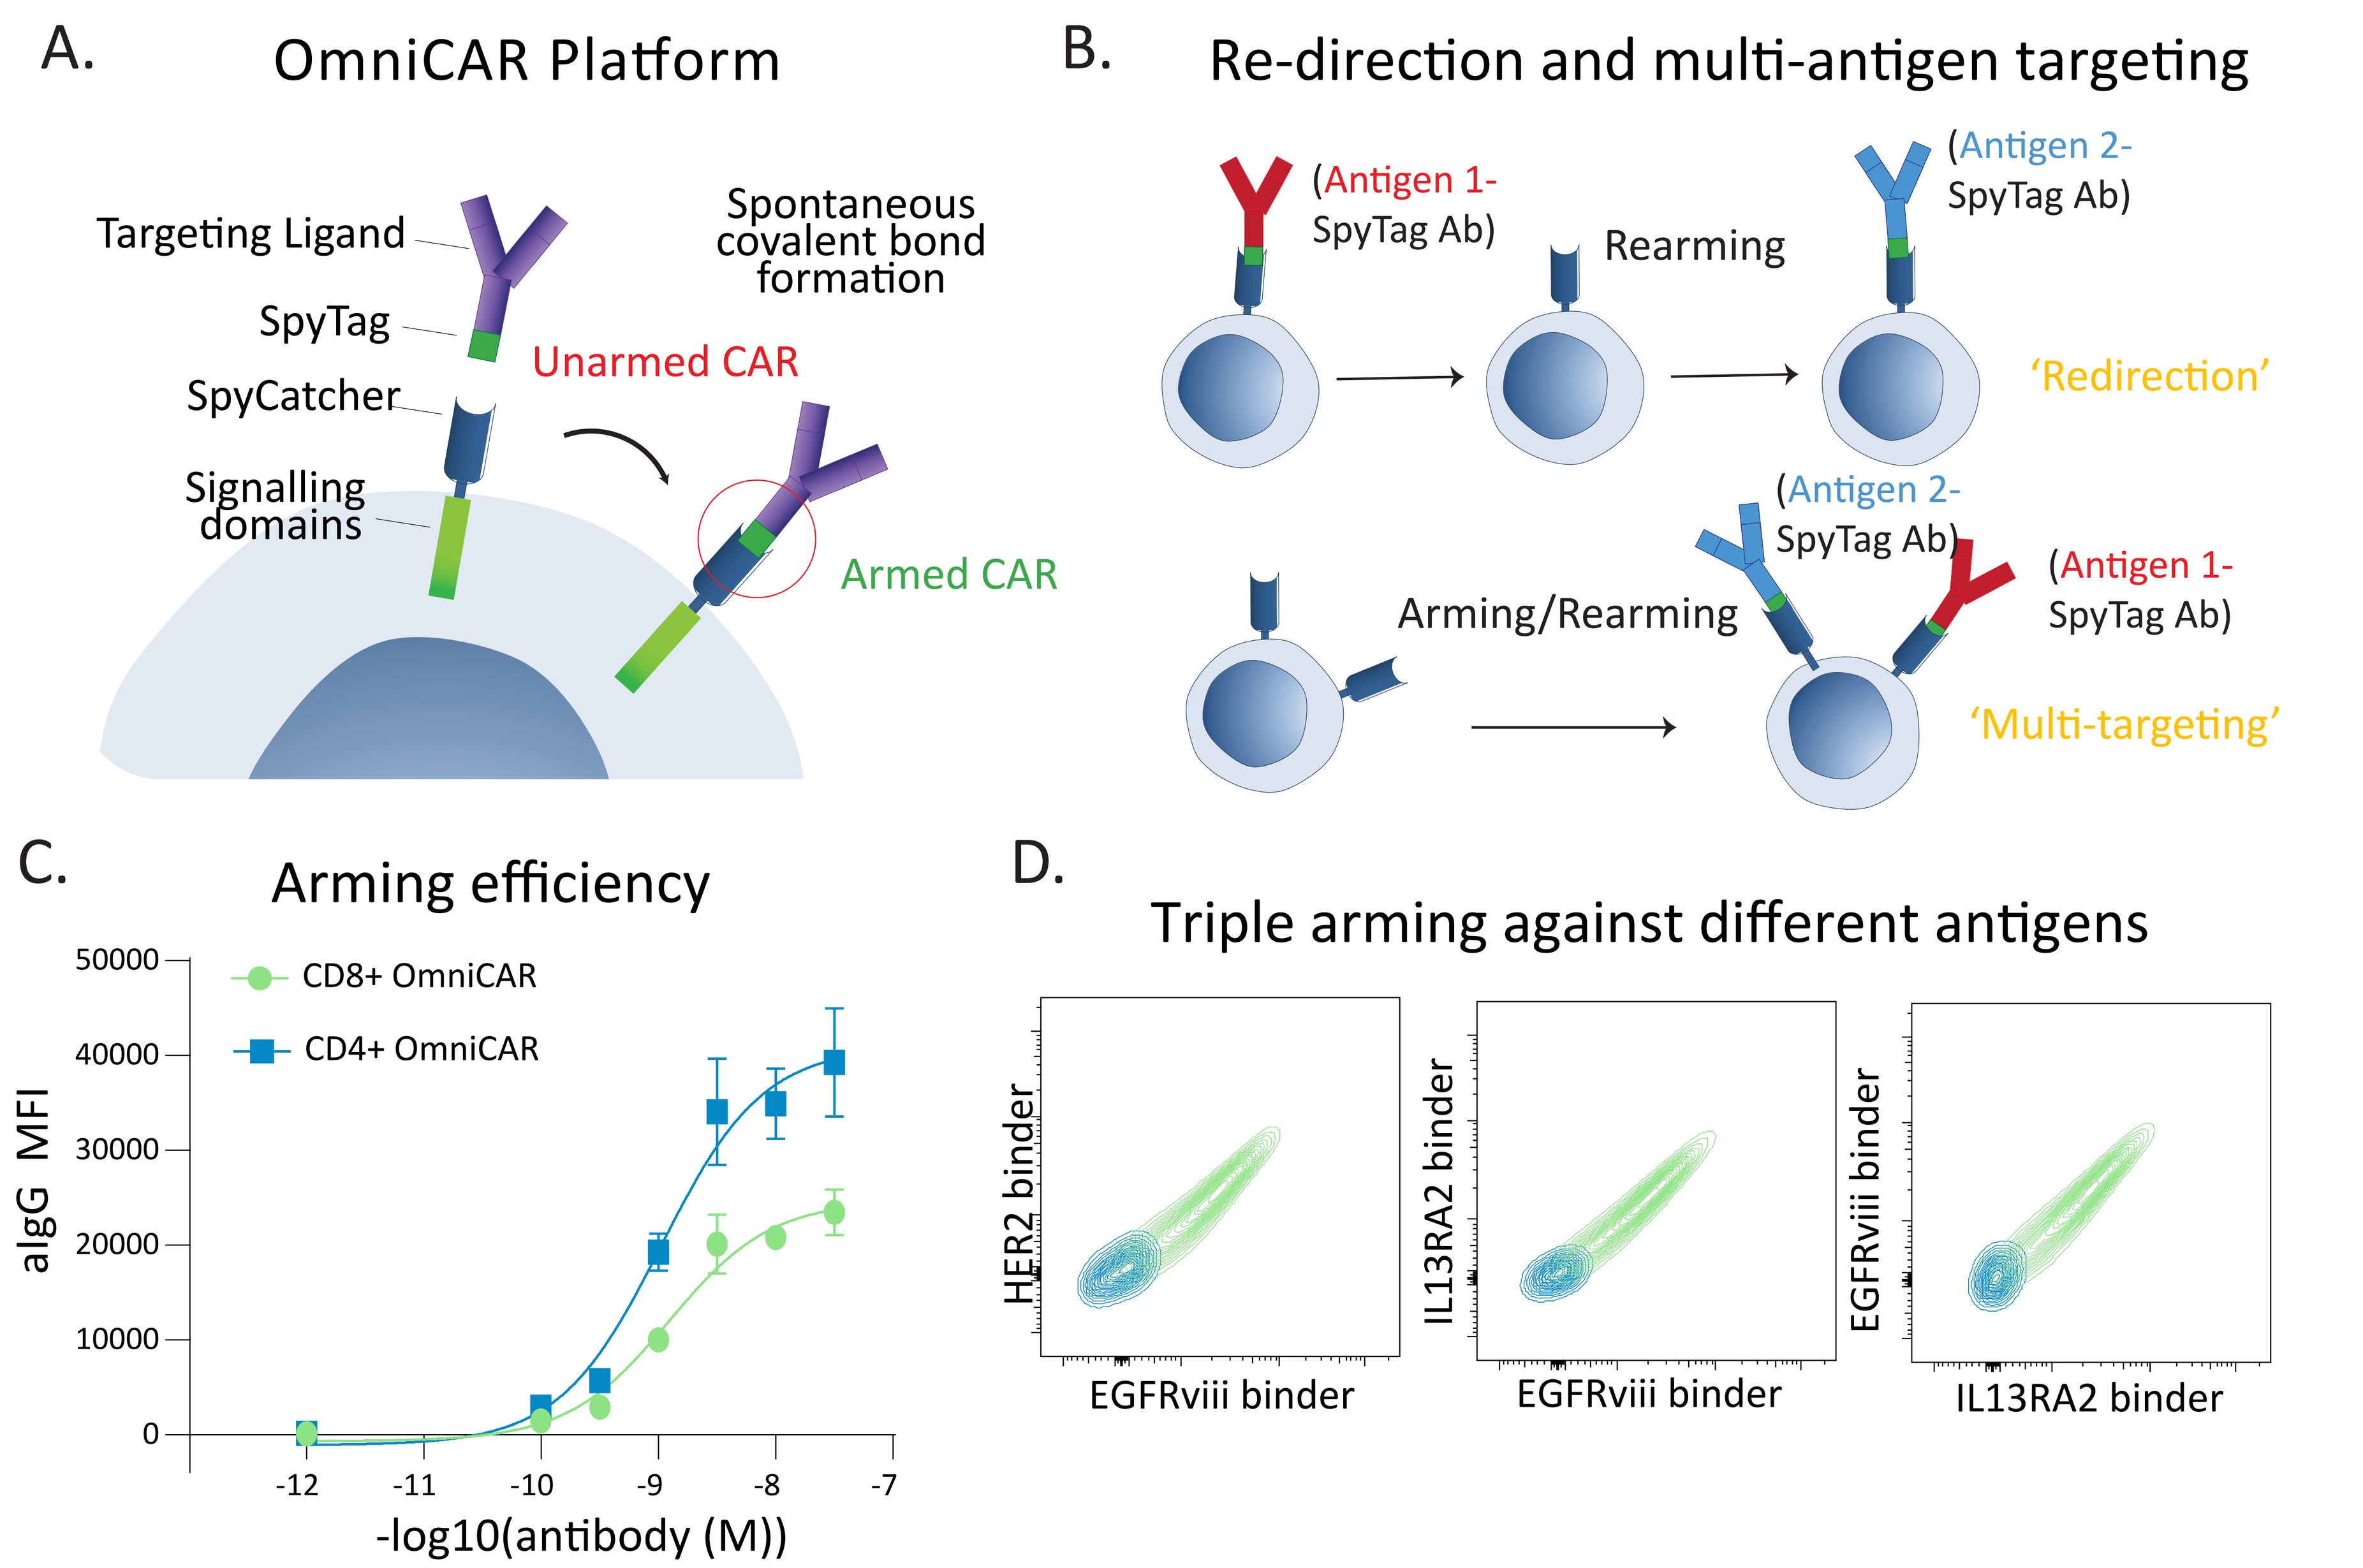
A.
OmniCAR Platform
Targeting Ligand
SpyTag
SpyCatcher
Signalling domains
Spontaneous covalent bond formation
Unarmed CAR
Armed CAR
B.
Re-direction and multi-antigen targeting
(Antigen 1-SpyTag Ab)
Rearming
(Antigen 2-SpyTag Ab)
'Redirection'
Arming/Rearming
'Multi-targeting'
C.
Arming efficiency
CD8+ OmniCAR
CD4+ OmniCAR
aIgG MFI
-log10(antibody (M))
D.
Triple arming against different antigens
HER2 binder
EGFRviii binder
IL13RA2 binder

Figure 1. OmniCAR can be armed to target multiple tumor antigens through spontaneous and rapid SpyCatcher/SpyTag covalent bond formation. (A) Schematic of the OmniCAR platform. (B) OmniCAR T cells can be 'armed' with different antigen targeting ligands prior to or post adoptive transfer, to redirect anti-tumor function against a different antigen target, or to target multiple antigens simultaneously. (C) Titratable arming efficiency in CD8+ and CD4+ OmniCAR T cells. (D) Triple arming against HER2, EGFRviii and IL13RA2 antigens

Precise control of anti-tumor function and killing by OmniCAR T cells against multiple different solid and liquid tumors

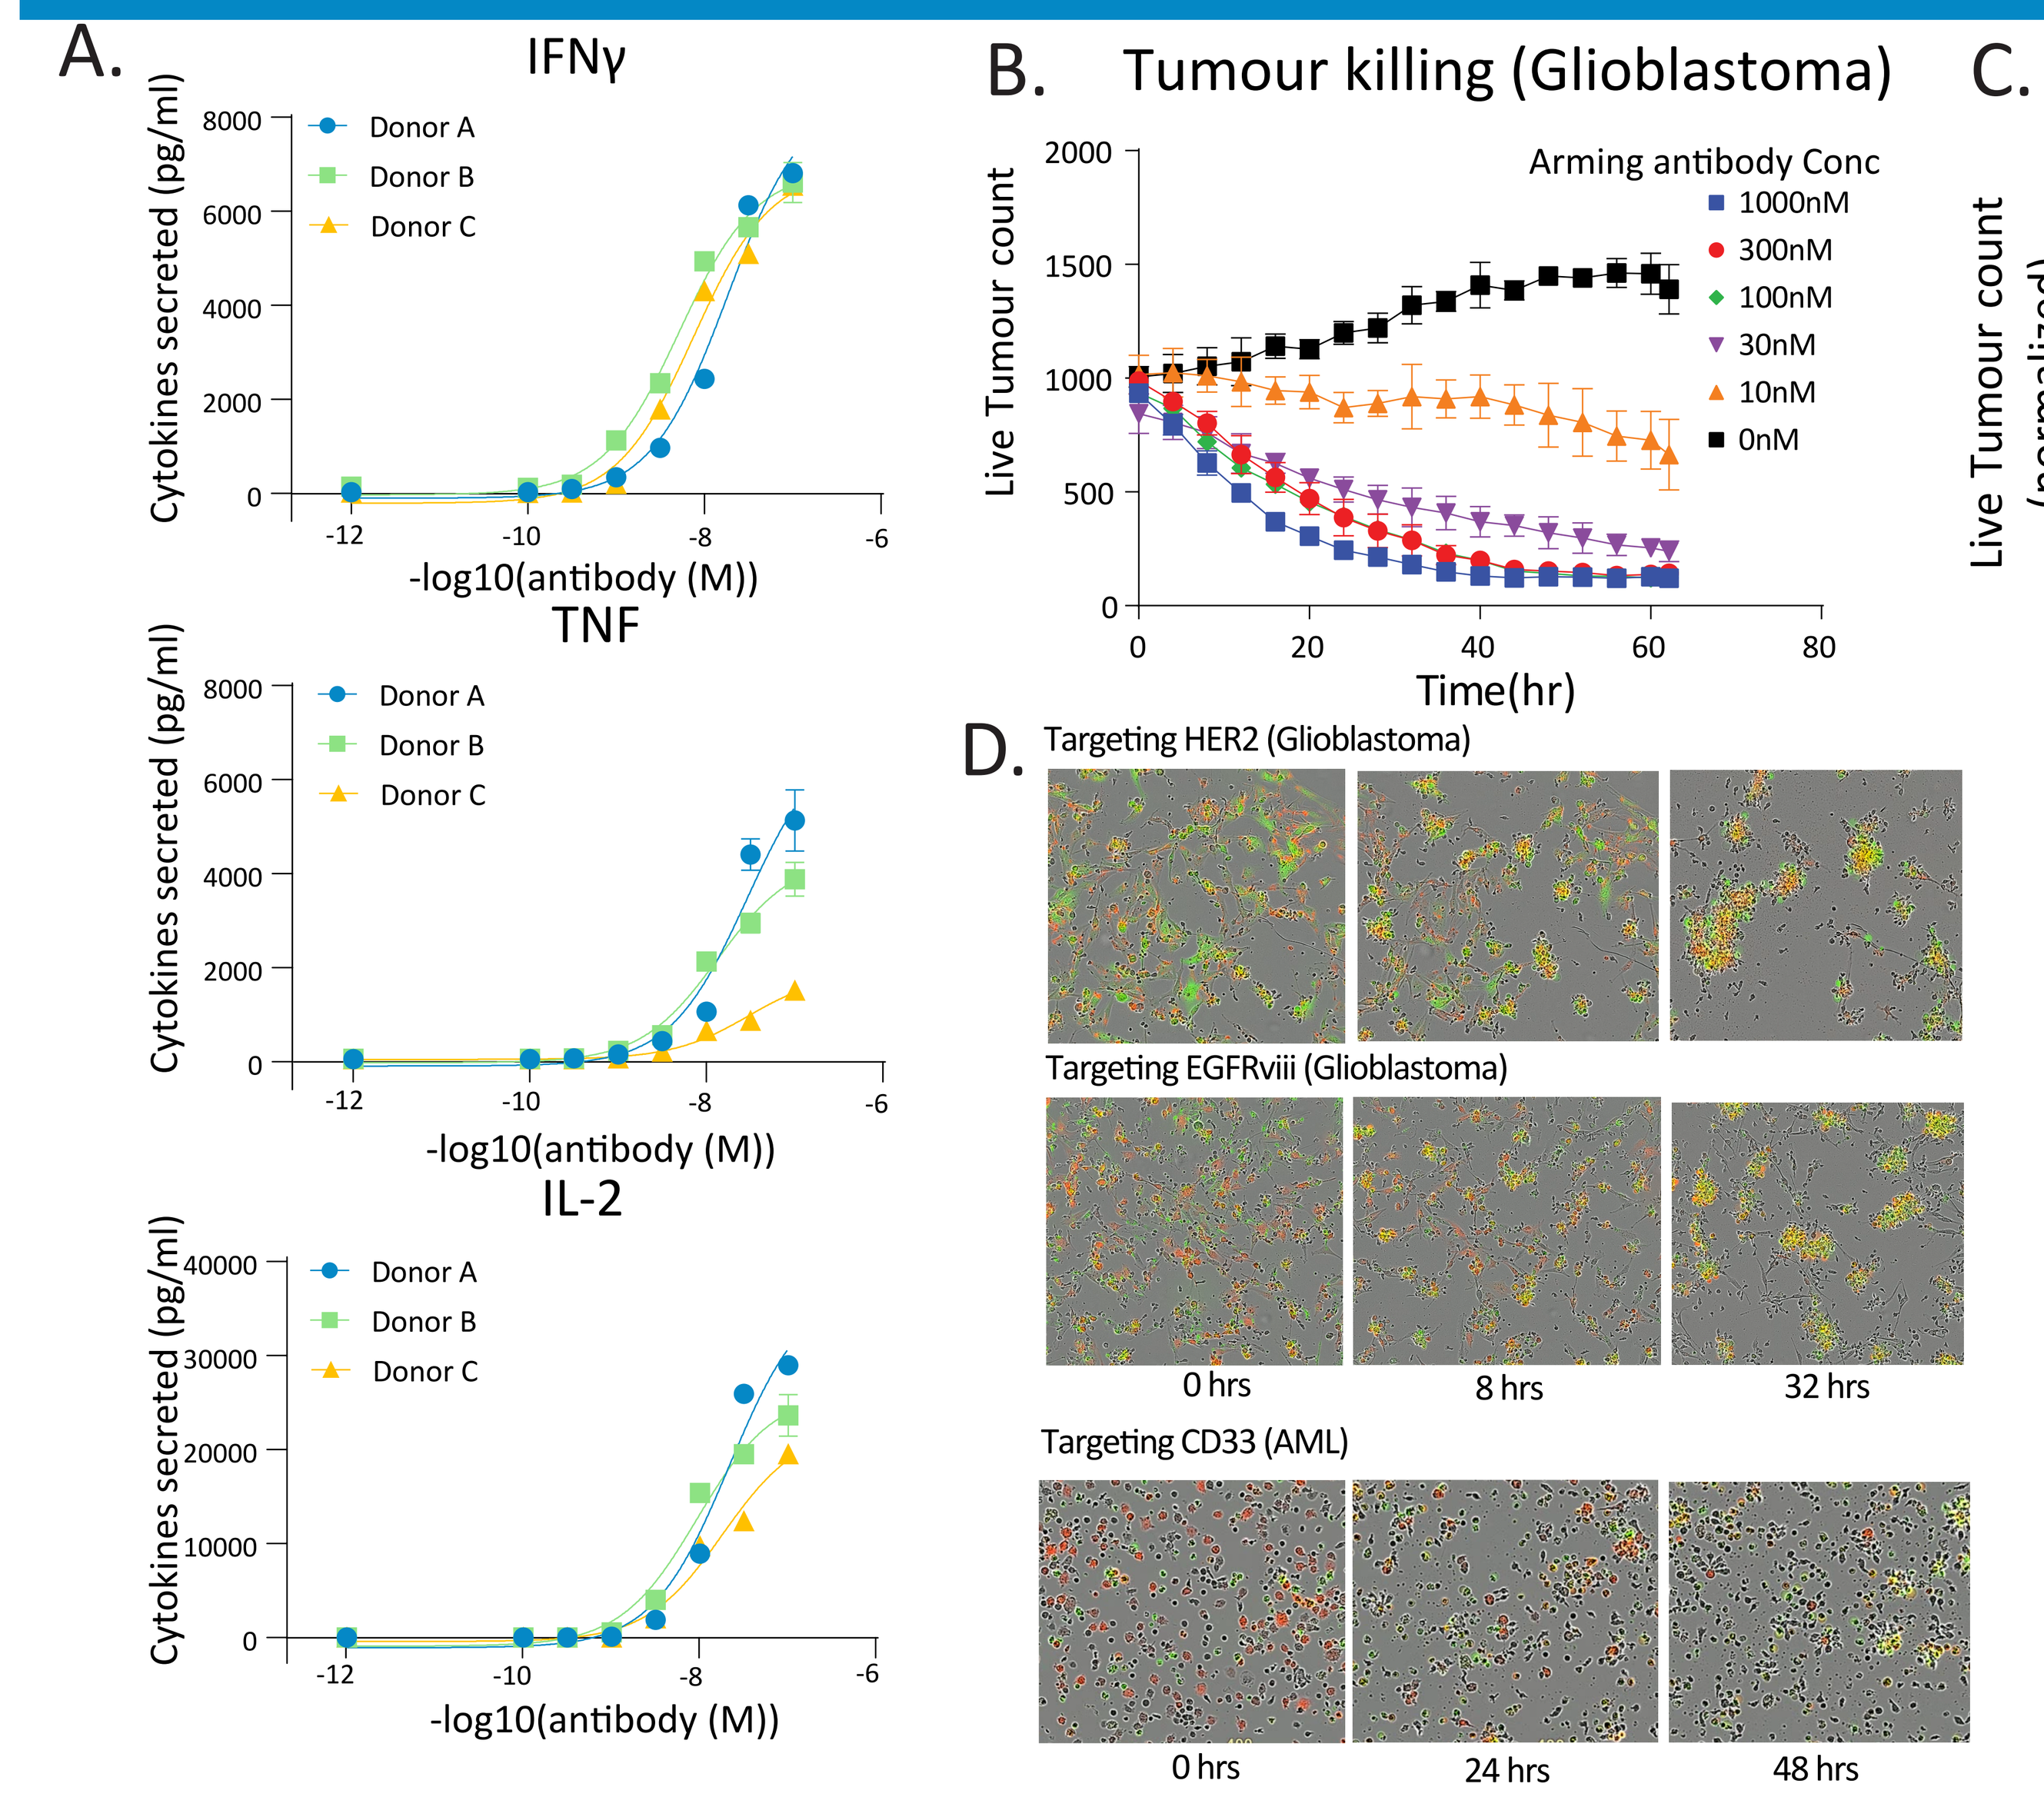
A.
IFNγ
TNF
IL-2
Donor A
Donor B
Donor C
Cytokines secreted (pg/ml)
-log10(antibody (M))
B.
Tumour killing (Glioblastoma)
Arming antibody Conc
1000nM
300nM
100nM
30nM
10nM
0nM
Live Tumour count
Time(hr)
C.
Tumour killing (AML)
CD33 armed
Unarmed
Tumour alone
Non-Transduced
Live Tumour count (normalized)
Time(hr)
D.
Targeting HER2 (Glioblastoma)
Targeting EGFRviii (Glioblastoma)
0 hrs
8 hrs
32 hrs
Targeting CD33 (AML)
0 hrs
24 hrs
48 hrs

Figure 2. Armed OmniCAR can effectively target multiple tumor antigens and solid and liquid tumor types with titratable precision. (A) Titratable cytokine production after overnight coculture of anti-HER2 binder armed OmniCAR T cells with MDA-MB231-HER2 breast cancer cells. (B) Titratable tumor killing of U251MG-EGFRviii glioblastoma with OmniCAR T cells armed with variable concentrations of anti-EGFRviii binders. (C) Targeting AML (MV411) cell lines with CD33-armed OmniCAR T cells (D) Incucyte snapshots of killing at different timepoints for solid and liquid tumors. Green (Caspase 3/7 cleavage), Red (Tumor cells), Unstained (OmniCAR T cells)

Specific activity and targeted re-direction of OmniCAR killing activity in mixed coculture of GBM cells expressing HER2 or EGFRviii

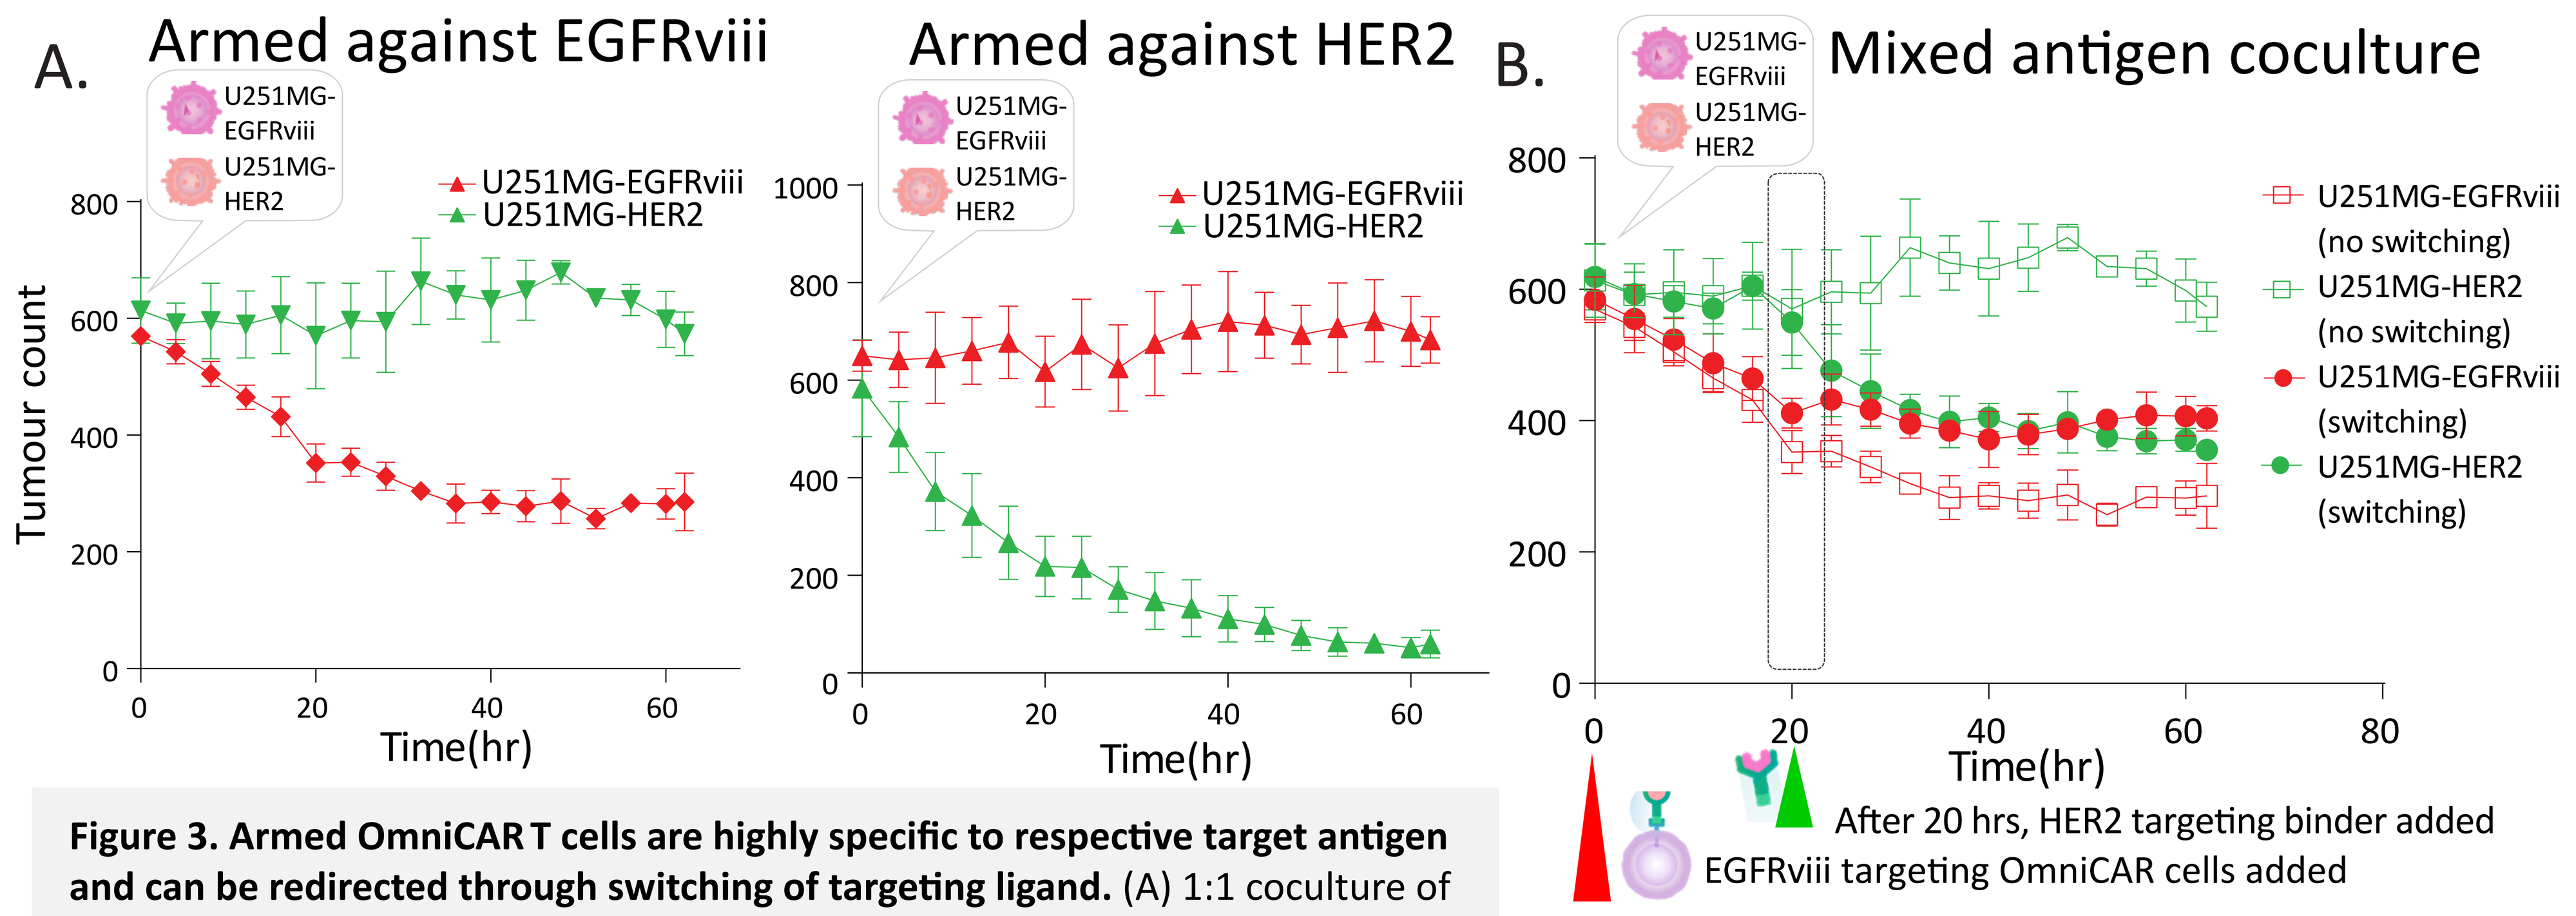
A.
Armed against EGFRviii
Armed against HER2
U251MG-EGFRviii
U251MG-HER2
Tumour count
Time(hr)
B.
Mixed antigen coculture
U251MG-EGFRviii (no switching)
U251MG-HER2 (no switching)
U251MG-EGFRviii (switching)
U251MG-HER2 (switching)
Time(hr)
After 20 hrs, HER2 targeting binder added
EGFRviii targeting OmniCAR cells added

Figure 3. Armed OmniCAR T cells are highly specific to respective target antigen and can be redirected through switching of targeting ligand. (A) 1:1 coculture of mixed EGFRviii or HER2 expressing U251 glioblastoma tumor cells is cultured with OmniCAR T cells armed with EGFRviii or HER2 binders at a 1:1 E:T ratio. (B) Mixed coculture with EGFRviii targeting OmniCAR T cells where anti-HER2 binders are only added at 20hrs to demonstrate sequential arming and killing.

Phenotyping of adoptively transferred OmniCAR T cells dosed with targeting binders in vivo demonstrates optimal dose for engraftment and formation of T-central memory compartment

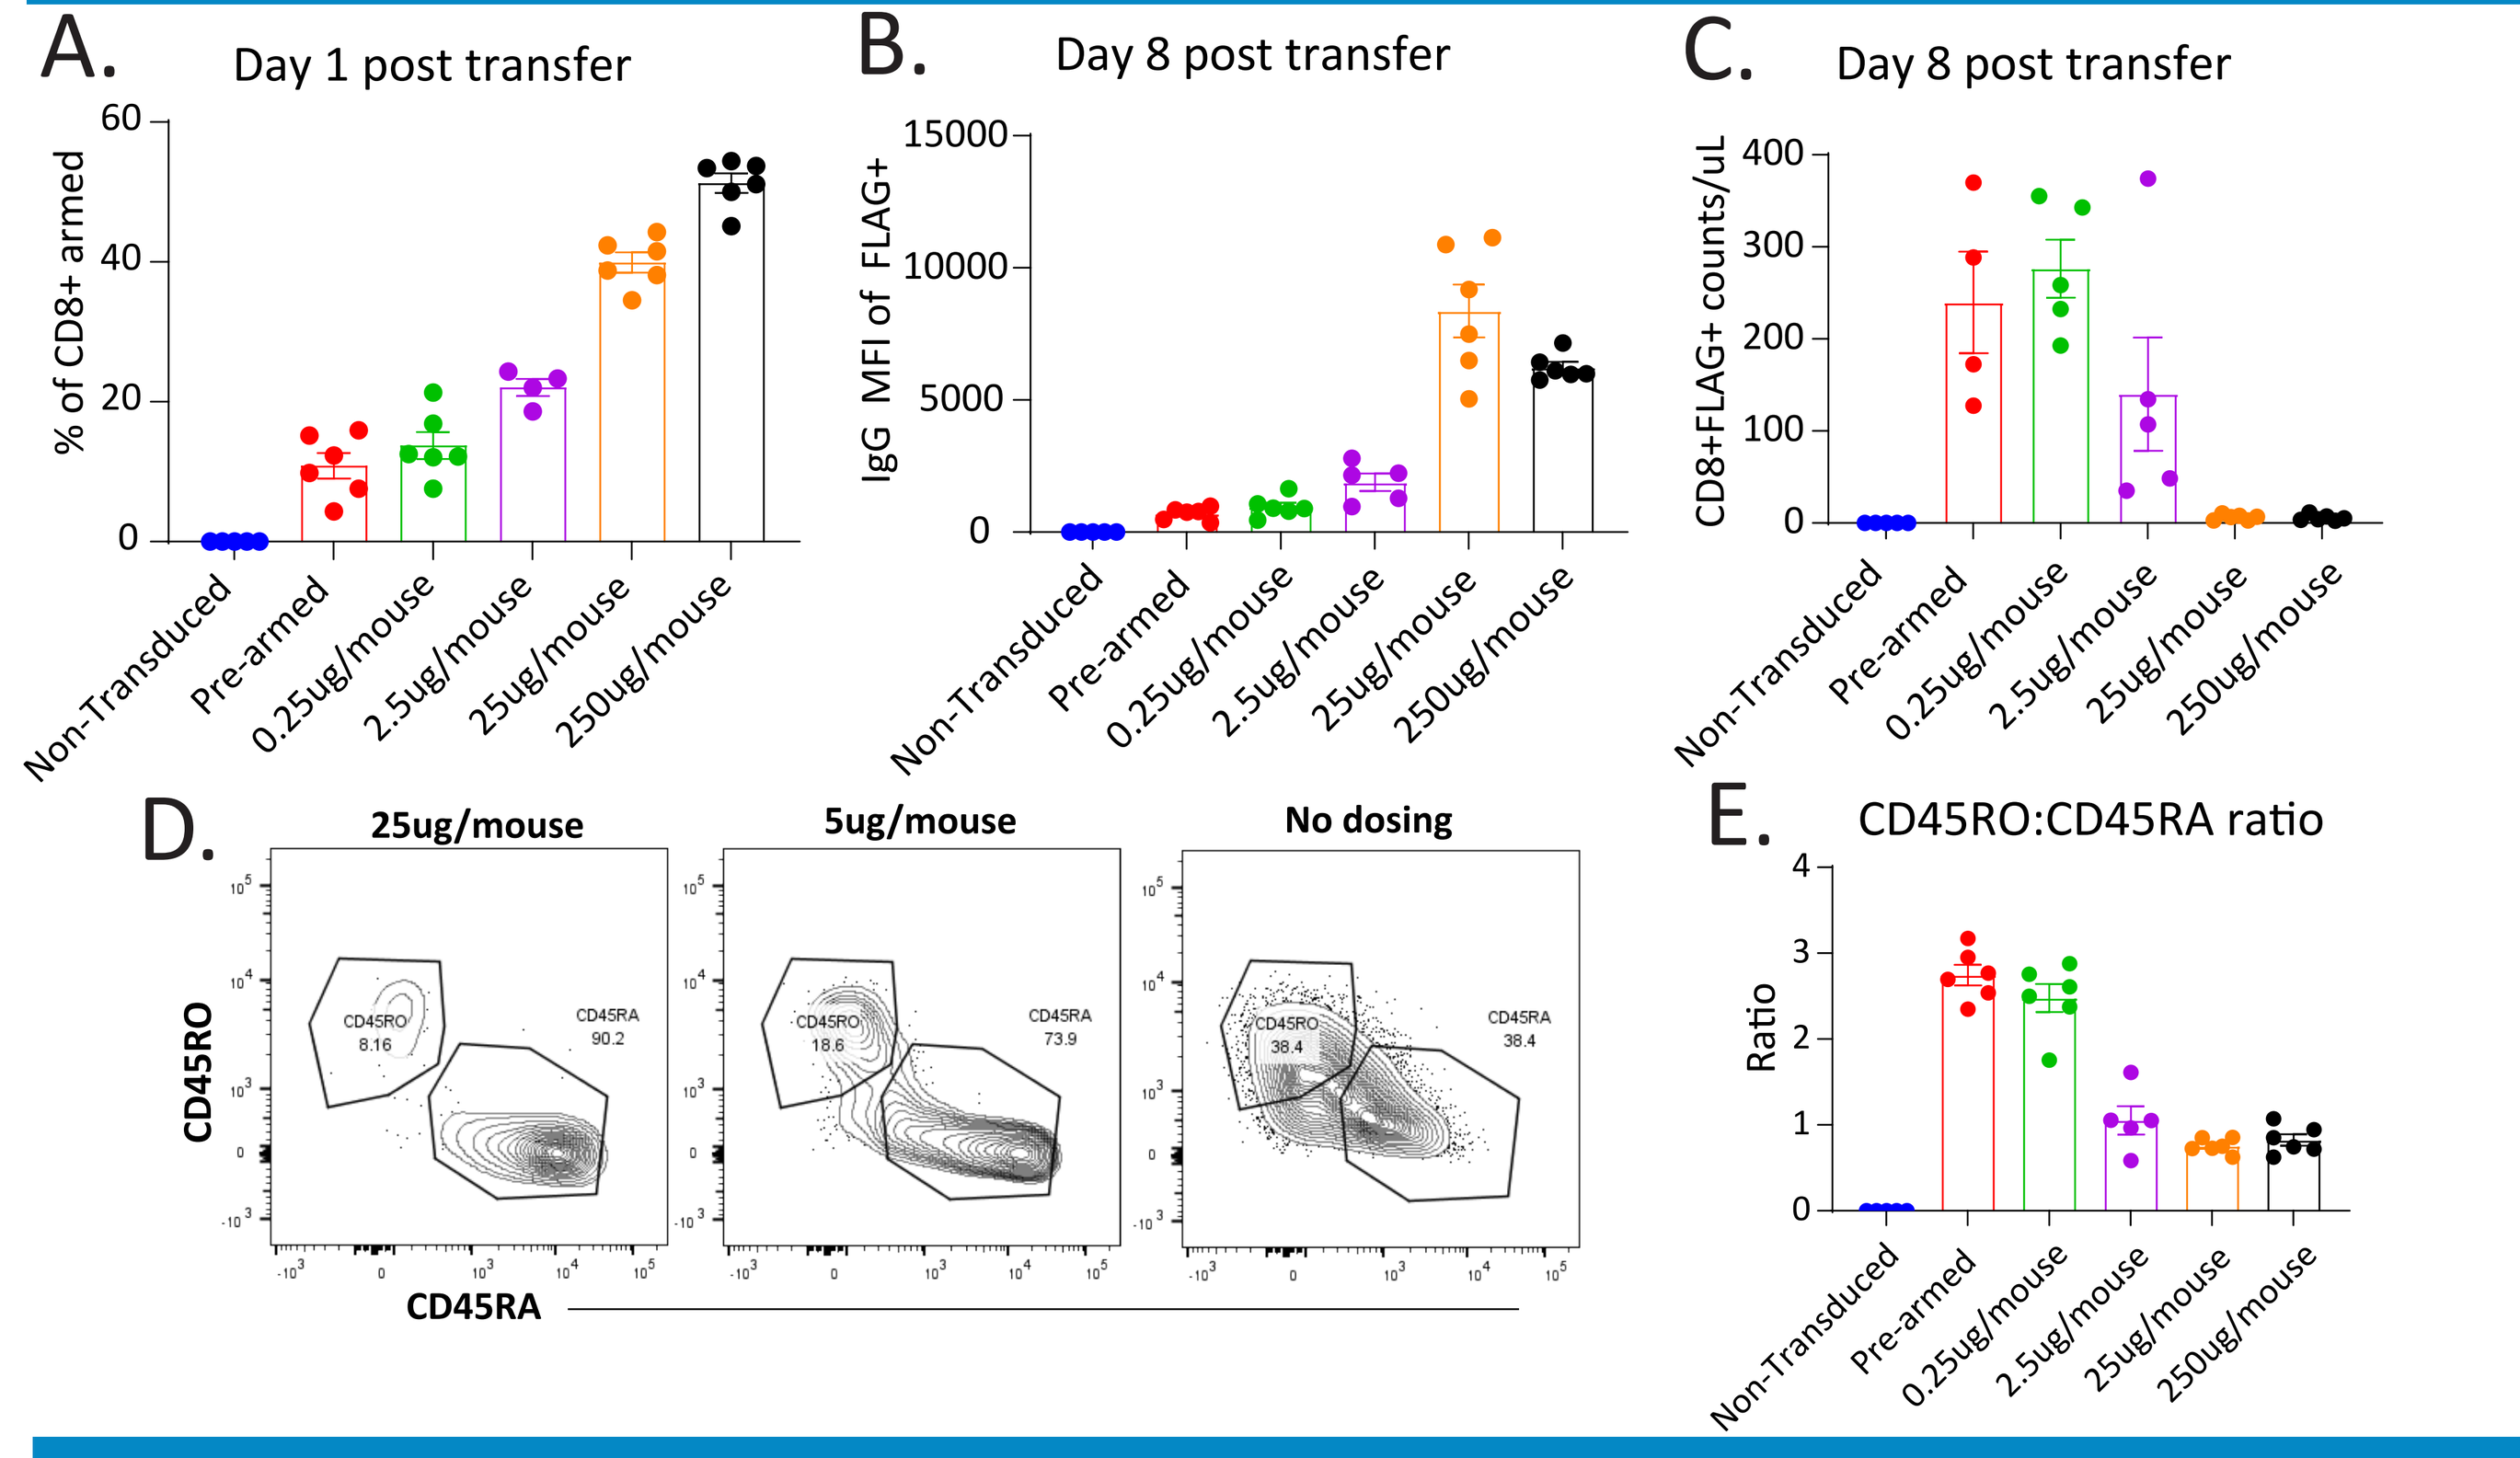
A.
Day 1 post transfer
% of CD8+ armed
B.
Day 8 post transfer
IgG MFI of FLAG+
C.
Day 8 post transfer
CD8+FLAG+ counts/uL
Non-Transduced
Pre-armed
0.25ug/mouse
2.5ug/mouse
25ug/mouse
250ug/mouse
D.
25ug/mouse
5ug/mouse
No dosing
CD45RO
CD45RA
E.
CD45RO:CD45RA ratio
Ratio

Figure 4. Titratable arming in vivo of adoptively transferred OmniCAR T cells when dosed with antibody binders I.P. 7.5e6 OmniCAR T cells was transferred into MDA-MB231-HER2 tumor bearing NSG mice. Immediately post transfer, mice to receive binders were dosed I.P. Circulating OmniCAR T cells in blood at (A) day 1 or (B) day 8 post transfer. (C) Quantification of OmniCAR numbers in blood. (D) Memory phenotype and (E) quantification of CD45RO:CD45RA ratio.

Resting OmniCAR T cells prior to dosing of targeting binders, drives potent anti-tumor efficacy against breast cancer in a xenograft model

Figure 5. Resting OmniCAR cells post adoptive transfer prior to dosing with antibody binders promote engraftment, expansion and potent anti-tumor function against solid tumors. (A) MDA-MB231-HER2 breast cancer bearing NSG mice were treated intrabenously with 7.5e6 unarmed OmniCAR T cells. After 21 days post adoptive transfer, anti-HER2 binders (25ug/mouse I.P) was dosed every 2-3 days until day 30 of experiment. (B) Combined tumor growth curves and (C) Individual tumor growth curves.

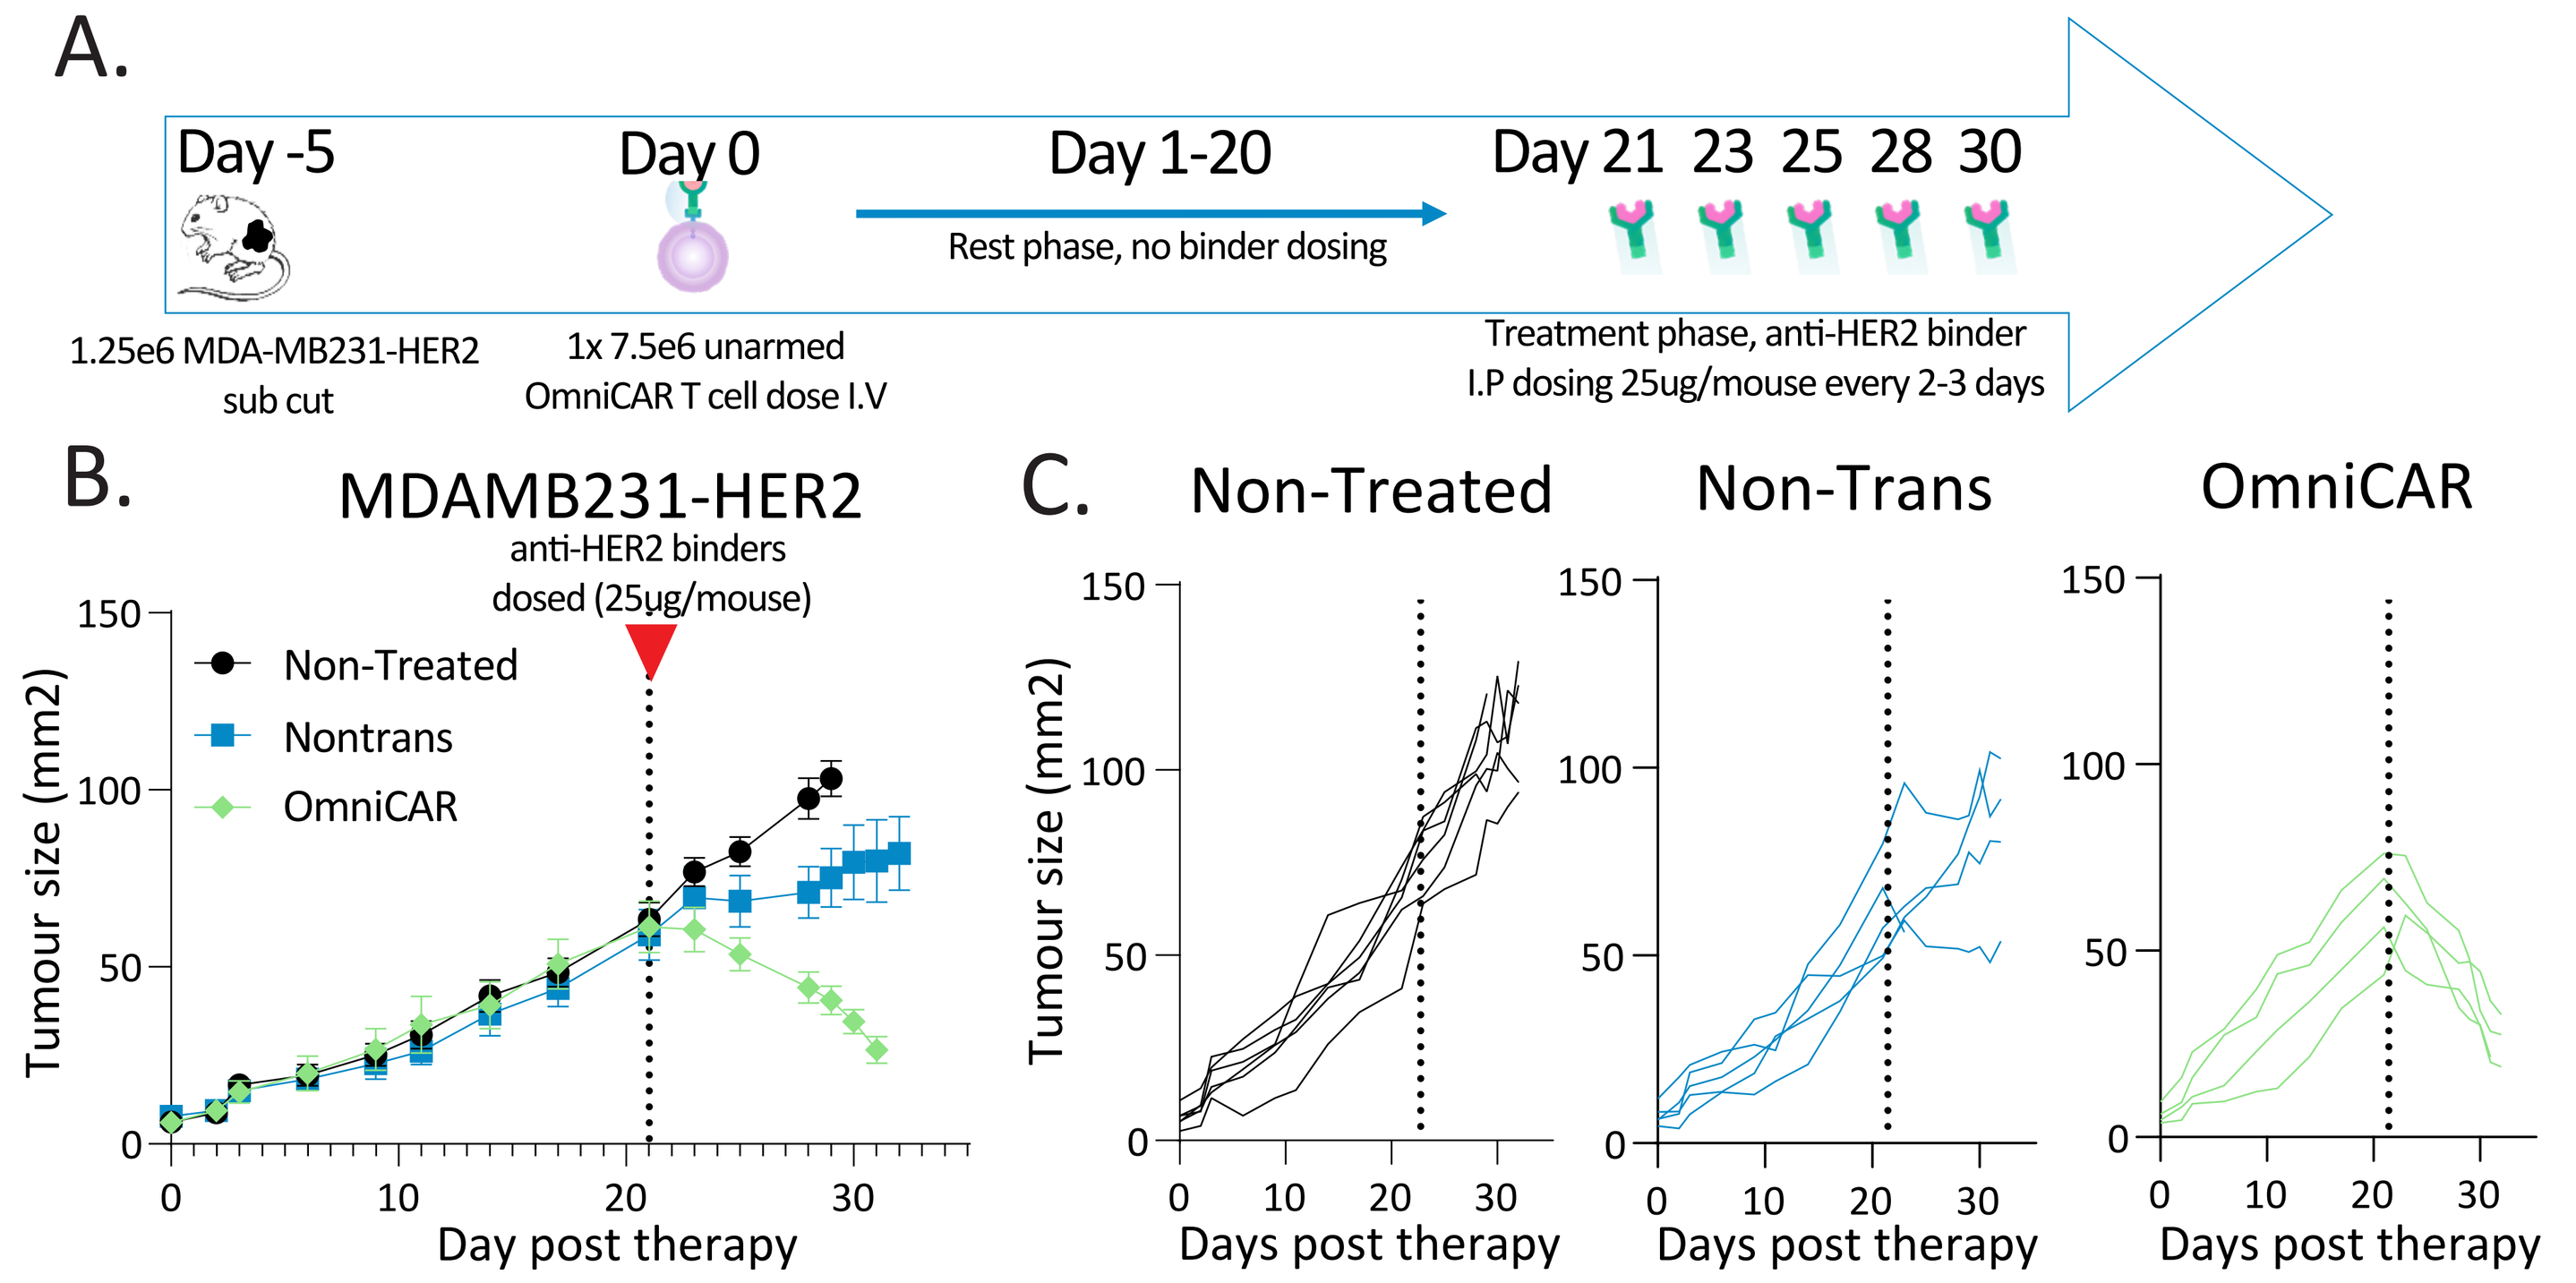
A.
Day -5
Day 0
Day 1-20
Day 21 23 25 28 30
Rest phase, no binder dosing
1.25e6 MDA-MB231-HER2 sub cut
1x 7.5e6 unarmed OmniCAR T cell dose I.V
Treatment phase, anti-HER2 binder I.P dosing 25ug/mouse every 2-3 days
B.
MDAMB231-HER2
anti-HER2 binders dosed (25μg/mouse)
Non-Treated
Nontrans
OmniCAR
Tumour size (mm2)
Day post therapy
C.
Non-Treated
Non-Trans
OmniCAR
Tumour size (mm2)
Days post therapy

Conclusions

OmniCAR is a universal CAR T cell platform that leverages the flexibility and versatility of antibody binders and spontaneous SpyCatcher/SpyTag covalent bond formation to target multiple antigens and tumor types. The precision afforded through dose-response 'arming' allows for titratable effector function to address toxicities post-transfer. The ability to target multiple antigens simultaneously or re-direct to a second antigen addresses tumor escape and heterogeneity. Finally, we also demonstrate that transferring unarmed OmniCAR T cells and introducing a 'rest' phase prior to arming leads to reduced exhaustion, better engraftment and memory phenotype and more potent anti-tumor efficacies.